P102

MRI, Safety, and Efficacy Outcomes in Patients with Relapsing MS: 18-month Results from the Long-term Extension Study of Tolebrutinib

Jiwon Oh[1], Robert J. Fox[2], Douglas L. Arnold[3,4], Sana Syed[5], Larry Orogun[5], Deborah Dukovic[6], Timothy Turner[5], Anthony Traboulsee[7], Daniel S. Reich[8]

[1]St. Michael's Hospital, University of Toronto, Toronto, ON, Canada; [2]Mellen Center for Multiple Sclerosis, Cleveland Clinic, Cleveland, OH, USA; [3]McGill University, Montréal, QC, Canada; [4]NeuroRx Research, Montréal, QC, Canada; [5]Sanofi, Cambridge, MA, USA; [6]Sanofi, Bridgewater, NJ, USA; [7]University of British Columbia, Vancouver, BC, Canada; [8]National Institute of Neurological Disorders and Stroke, Bethesda, MD, USA

OBJECTIVE

- **To report magnetic resonance imaging (MRI), safety, and efficacy outcomes at Week (W) 72 (Month 18) of the tolebrutinib long-term safety (LTS) extension of the phase 2b tolebrutinib trial in patients with relapsing multiple sclerosis (MS)**

INTRODUCTION

- Tolebrutinib is an oral, brain-penetrant inhibitor of Bruton's tyrosine kinase (BTK) currently under investigation as a potential treatment for MS[1]
- BTK inhibition by tolebrutinib results in modulation of B-cell function rather than depletion[2,3]
- Tolebrutinib also inhibits Fcγ receptor-dependent signalling in myeloid cells, including microglia, through durable occupancy of BTK[1]
- In a dose-finding phase 2b trial (NCT03889639 [DRI15928]), tolebrutinib 60 mg daily over 12 weeks reduced formation of new contrast-enhancing lesions and new or enlarging T2 lesions by >85% versus placebo in patients with relapsing MS (RMS)[4]
- Tolebrutinib was well tolerated over 12 weeks, and safety was consistent across doses
- The 60-mg dose is being tested in the phase 3 trials GEMINI 1 (NCT04410978) and GEMINI 2 (NCT04410991) for RMS, and PERSEUS (NCT04458051) and HERCULES (NCT04411641) for progressive MS,[5] all of which are currently recruiting
- LTS16004 (NCT03996291) is an ongoing long-term extension study of tolebrutinib in patients who completed the phase 2b study

METHODS

Study Design

- Patients who completed the double-blind period (DBP) were eligible to enrol in the LTS study Part A, where they continued (in a double-blinded manner) the tolebrutinib dose from the core study (5, 15, 30, or 60 mg/day; **Figure 1**)
- Once the 60-mg dose was selected for the phase 3 trials, patients entered the open-label LTS study Part B, in which they all receive tolebrutinib 60 mg/day

Figure 1. Study Design

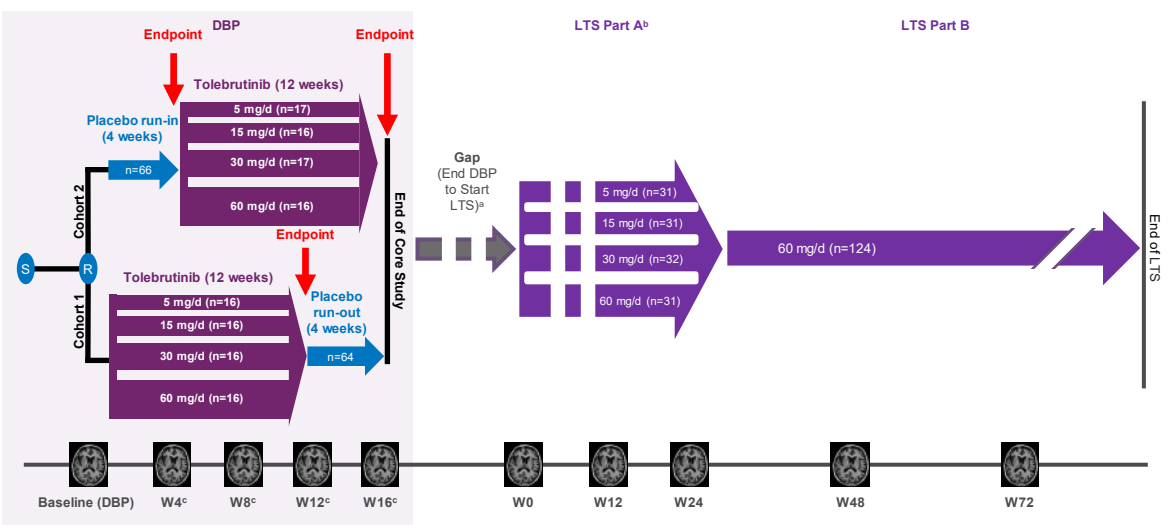
DBP
Endpoint
Endpoint
LTS Part A[b]
LTS Part B
Tolebrutinib (12 weeks)
Placebo run-in (4 weeks)
n=66
5 mg/d (n=17)
15 mg/d (n=16)
30 mg/d (n=17)
60 mg/d (n=16)
Cohort 2
Cohort 1
S
R
Tolebrutinib (12 weeks)
5 mg/d (n=16)
15 mg/d (n=16)
30 mg/d (n=16)
60 mg/d (n=16)
Placebo run-out (4 weeks)
n=64
End of Core Study
Gap (End DBP to Start LTS)[a]
5 mg/d (n=31)
15 mg/d (n=31)
30 mg/d (n=32)
60 mg/d (n=31)
60 mg/d (n=124)
End of LTS
Baseline (DBP)
W4[c]
W8[c]
W12[c]
W16[c]
W0
W12
W24
W48
W72

[a]Gap period in the transition between last dose in the DBP and first dose in the LTS study was variable (mean ± SD, 7 ± 7.3 weeks; range, 0–21 weeks). [b]Duration of Part A of the LTS study was variable (mean ± SD, 27.4 ± 6.3 weeks; range, 15–47 weeks). [c]DBP MRI scans in this figure are labelled according to the week of the DBP; in contrast, the DBP time points in Figures 2 and 3 indicate the number of weeks on tolebrutinib treatment in the DBP.
DBP=double-blind period; LTS=long-term safety; R=randomization; S=screening; W=Week.

References

[1]Smith PF, et al. ACTRIMS 2019, Poster 072. [2]Sanofi Genzyme. Data on file. [3]Reich D, et al. EAN 2020, Presentation O4010. [4]Reich DS, et al. *Lancet Neurol* 2021;20:729-38. [5]Vermersch P, et al. EAN 2021, Poster EPO-455. [6]Elliott C, et al. *Mult Scler J* 2018;25:1915-25.

CONCLUSIONS

- **Nearly all patients (94%) who enrolled in the long-term extension study of tolebrutinib in patients with RMS have remained on study as of September 6, 2021 (18-month cut-off)**
- **New Gd-enhancing lesion counts remained low for the tolebrutinib 60-mg arm and were reduced in lower dose arms by the LTS study W48/72 when all patients had switched to 60 mg**
- **Safety data continue to show favourable tolerability without the emergence of any new safety signals**
- **ARR in patients on tolebrutinib 60 mg was low, and ~85% of patients were free of relapses. EDSS scores remained stable**
- **Longer follow-up in the ongoing extension, as well as data from the phase 3 trials, will continue to build on the safety and efficacy profile of tolebrutinib in patients with MS**

Please scan to download the poster

Outcomes

- MRI outcomes included the number of new gadolinium (Gd)-enhancing T1 lesions, number of new or enlarging T2 lesions, T2 lesion volume change from baseline, volume of slowly evolving lesions (SELs) at the LTS study W72, and number of paramagnetic rim lesions (PRLs) at LTS W72
- Gd-enhancing lesions were identified by ≥1 radiologist masked to treatment assignment at an independent, central facility (NeuroRx, Montréal, QC, Canada)
- New/enlarging T2 lesions were identified from sequential scans using a semi-automated process of joint timepoint segmentation, followed by manual correction as necessary
- SELs were identified as contiguous areas of the baseline T2-lesion mask that showed constant and concentric local expansion from baseline to W72, with local expansion determined by Jacobian analysis[6]
- PRLs were manually identified on susceptibility-weighted images generated from 6 3D-gradient echo phase images in a subset of patients at sites with sufficient imaging capability
- Safety includes the percentage of participants with treatment-emergent adverse events (TEAEs)
- Clinical efficacy outcomes:
 – Annualized relapse rate (ARR) and relapse frequency, evaluated in all patients who received at least 8 weeks of treatment with tolebrutinib 60 mg in the LTS extension study up to the W72 cut-off
 – Mean Expanded Disability Status Scale (EDSS) score over the LTS extension study by treatment group

RESULTS

Patients

- Of 129 eligible participants, 125 continued in the long-term extension study, and 124 completed Part A and transitioned to Part B
- As of September 6, 2021 (W72 cut-off), 118 (94%) participants remained on study, with median time 21.4 months
- At DBP baseline, the mean ± SD age of enrolled patients was 37.7 ± 9.6 years (range 19-56); 69% were women

Gd-enhancing Lesion Counts

- Numbers of new Gd-enhancing lesions remained low in the 60/60-mg arm through W72 and were reduced in other arms by W48 and W72 when all patients had switched to 60 mg (**Figure 2**)

Figure 2. Gd-Enhancing Lesion Counts

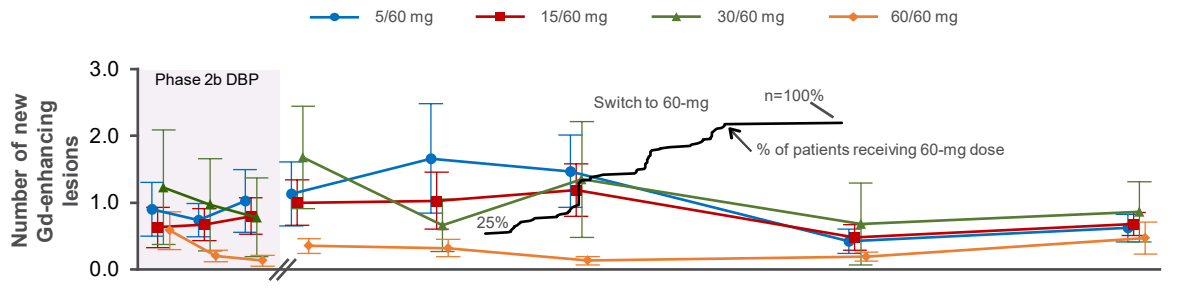
5/60 mg
15/60 mg
30/60 mg
60/60 mg
Number of new Gd-enhancing lesions
Phase 2b DBP
Switch to 60-mg
n=100%
% of patients receiving 60-mg dose
25%
W4 W8 W12 W0 W12 W24 W48 W72
DBP
LTS Part A/B

No. of Patients:	W4	W8	W12	W0	W12	W24	W48	W72
5/60 mg	30	31	30	31	29	30	26	26
15/60 mg	30	30	30	31	29	31	31	31
30/60 mg	31	31	32	31	32	31	31	29
60/60 mg	31	30	30	31	28	31	31	30

Data are mean (SE). The switches to 60-mg from the lower doses occurred between W15-W47 (the black line shows the percentage of patients receiving the 60-mg dose).

Figure 3. T2 Lesion Volume Change

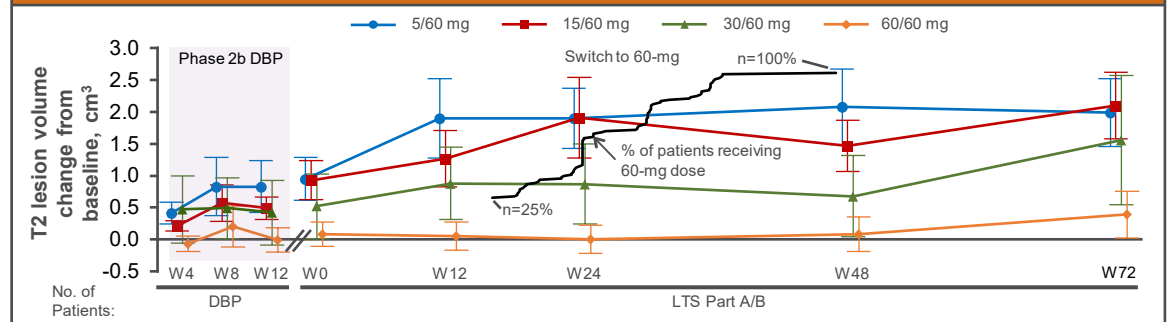
5/60 mg
15/60 mg
30/60 mg
60/60 mg
T2 lesion volume change from baseline, cm³
Phase 2b DBP
Switch to 60-mg
n=100%
% of patients receiving 60-mg dose
n=25%
W4 W8 W12 W0 W12 W24 W48 W72
DBP
LTS Part A/B

No. of Patients:	W4	W8	W12	W0	W12	W24	W48	W72
5/60 mg	30	31	31	31	30	30	28	27
15/60 mg	30	30	30	30	28	30	30	30
30/60 mg	32	32	32	32	32	31	32	29
60/60 mg	31	31	31	31	29	31	31	30

Data are mean (SE). The switches to 60-mg from the lower doses occurred between W15-W47 (the black line shows the percentage of patients receiving the 60-mg dose).

T2 Lesion Volume Change

- T2 lesion volume change remained low in the 60/60-mg arm (W72 vs. baseline [mean ± SE]: +0.39 ± 0.36 cm^3; **Figure 3**)

New/Enlarging T2 Lesion Counts

- New/enlarging T2 lesion counts remained low for 60/60-mg through W24, increasing slightly at W48 and W72 where the interval between MRI scans was longer than at previous time-points

SEL Volume and PRL Count

- Median (IQR) W72 SEL volume was:
 – 441 (69-630) mm^3 in the 5/60-mg arm
 – 468 (102-1317) mm^3 in the 15/60-mg arm
 – 675 (150-1230) mm^3 in the 30/60-mg arm
 – 284 (168-504) mm^3 in the 60/60-mg arm
- In patients who had scans at both DBP baseline and W72 (n=24), the number of PRLs remained unchanged except in: 1 patient who had 1 PRL at baseline that was absent at W72 and 2 had additional PRLs at W72 compared with baseline (1 had 2 more, 1 had 3 more)[§]

[§]These 2 patients both had new Gd-enhancing lesions.

Figure 4. ARR (A) and Relapse Frequency (B) up to the LTS Study Week 72 Cut-off[a]

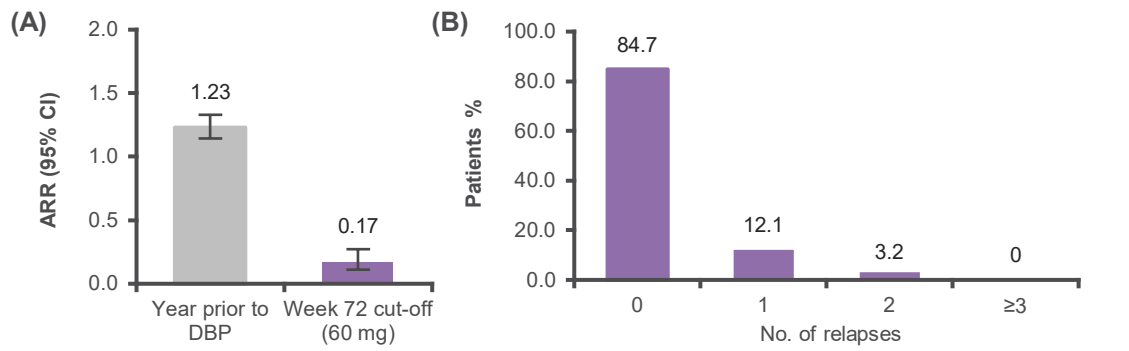
(A)
ARR (95% CI)
1.23
0.17
Year prior to DBP
Week 72 cut-off (60 mg)
(B)
Patients %
84.7
12.1
3.2
0
0
1
2
≥3
No. of relapses

[a]ARR after ≥8 weeks of tolebrutinib 60 mg treatment in LTS16004 (up to cut-off for analysis). For patients originally assigned to 5, 15, or 30 mg, only the patient years and relapses starting 8 weeks after the switch to Part B are included. For patients originally assigned to 60 mg, all LTS16004 data are included unless the sum of the DRI15928 placebo run-out period and any gap period to start of LTS16004 Part A was >4 weeks, in which case only the patient years and relapses starting 8 weeks after re-initiation of treatment are included.

Figure 5. Mean EDSS Score Over Time

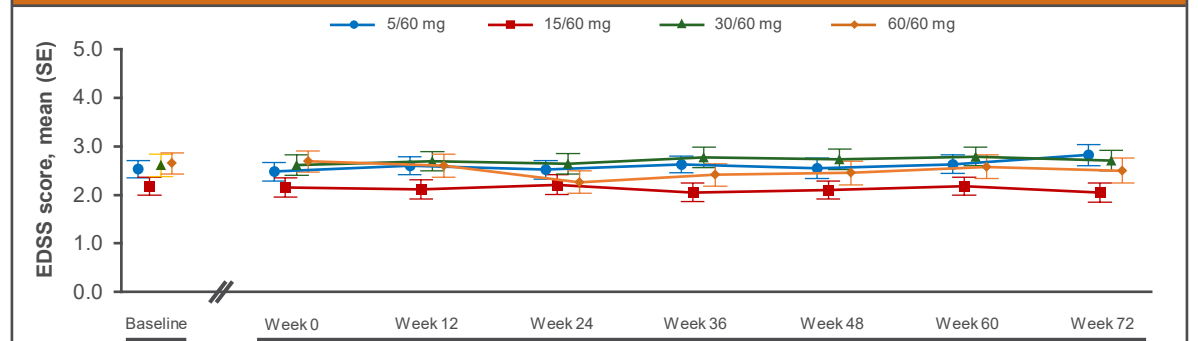
5/60 mg
15/60 mg
30/60 mg
60/60 mg
EDSS score, mean (SE)
Baseline (DBP)
Week 0 Week 12 Week 24 Week 36 Week 48 Week 60 Week 72
LTS Part A/B

No. of Patients:	Baseline (DBP)	Week 0	Week 12	Week 24	Week 36	Week 48	Week 60	Week 72
5/60 mg	31	31	31	29	27	28	28	28
15/60 mg	31	31	31	31	31	31	31	30
30/60 mg	32	32	32	27	32	31	31	31
60/60 mg	31	31	31	32	28	31	30	29

Clinical Outcomes

- ARR on tolebrutinib 60 mg was 0.17 (95% CI: 0.11, 0.27); 84.7% of patients remained relapse-free up to the LTS study W72 cut-off (**Figure 4**)
- Mean EDSS scores remained stable to LTS W72 (**Figure 5**)

Safety

- The most common TEAEs were COVID-19, headache, nasopharyngitis, upper respiratory tract infection, and arthralgia (**Table 1**)
- There was no suggestion of a dose effect for TEAEs or serious AEs in the LTS study Part A and no emergence of new safety signals for participants who switched to the 60-mg dose in Part B
- 1 patient (1%, on 5 mg/day) discontinued Part A due to progressive disease, and 6 discontinued Part B due to AEs (n=2 [2%])[†], perceived lack of efficacy (n=1 [1%]), progressive disease (n=1 [1%]), and emigration (n=2 [2%])

[†]AEs leading to treatment discontinuation were 1 headache and 1 alanine aminotransferase elevation.

Table 1. Most Common TEAEs Occurring in ≥5% of Patients[a]

TEAE	Patients, n (%)
COVID-19[b]	20 (16)
Headache	16 (13)
Nasopharyngitis	13 (10)
Upper respiratory tract infection	10 (8)
Arthralgia	7 (6)

[a]All patients (n=125). [b]All cases of COVID-19 were mild (n=11) or moderate (n=9) and resolved, and patients remained in the study. Three of the moderate COVID-19 cases were considered serious, of which 2 were hospitalized. Tolebrutinib treatment was interrupted temporarily in 4 patients. For details, see poster P126.

Acknowledgments and Disclosures

The authors and Sanofi thank the patients and their families for their involvement in the tolebrutinib phase 2b trial and its extension. This poster was reviewed by Svend S Geertsen, PhD, of Sanofi. Editorial support for the poster was provided by Richard J Hogan, PhD, and Gleb Baida, PhD, both of Elevate Scientific Solutions and was funded by Sanofi. JO: Consulting or speaking fees (Alexion, Biogen Idec, BMS, EMD Serono, Genzyme, Novartis, and Roche) and research support (Biogen Idec, EMD Serono, and Roche); RF: Consulting fees (Acorda, Bayer, Biogen, EMD Serono, Genentech, Novartis, Roche, Sanofi, Sanofi Genzyme, and Teva) and research support (Acorda, Biogen, Chugai, EMD Serono, Novartis, Opexa, Roche, Sanofi Genzyme, and Teva); DLA: Consulting fees (Albert Charitable Trust, Alexion Pharma, Biogen, Celgene, Frequency Therapeutics, Genentech, Med-Ex Learning, Merck, Novartis, Population Council, Receptos, Roche, and Sanofi-Aventis), grants (Biogen, Immunotec, and Novartis), and equity interest (NeuroRx); SS, LO, DD, TT: Employees of Sanofi (may hold shares and/or stock options in the company); AT: Consulting and/or speaking fees and grant/research support (Roche and Sanofi); DSR: Supported by the Intramural Research Program of NINDS, NIH. Additional research support (Abata, Sanofi, and Vertex).

Presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum, February 24–26, 2022, West Palm Beach, FL, USA
Funding provided by Sanofi